Machine Learning in Neuroplasticy

Michael J. Bower[1], Bhanushee Sharma[2], Christopher T. Lowden[2],
J. Christopher Culberson[2], and Milan Chytil[1]
1. Delix Therapeutics (Bedford, MA) 2. Workflow Informatics (Wake Forest, NC)

1. What are neuroplastogens?

Psychedelic compounds have demonstrated promise for treating neuropsychiatric disorders like depression and PTSD. The clinical effects of these compounds are hypothesized to be mediated by the promotion of rapid and enduring effects on structural neuroplasticity, including longer neurites, more dendritic spines, and more synapses. Delix calls molecules that cause one or more of these morphological changes neuroplastogens. Traditionally, this activity has been found in hallucinogenic natural products like LSD, psilocybin, ketamine, and ibogaine. Delix is researching non-hallucinogenic neuroplastogens to treat depression.

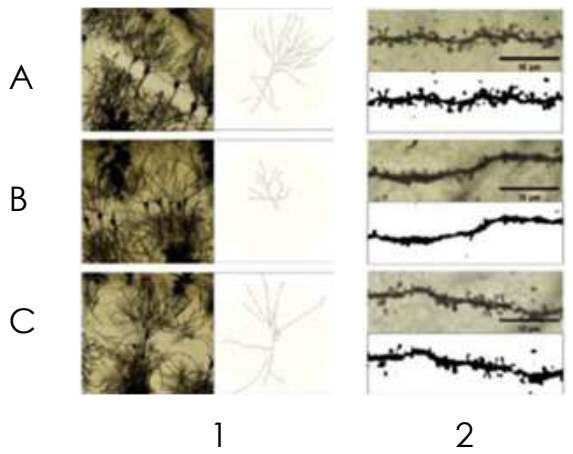
A
B
C
1
2

Representative images of 1) dendritic branches and 2) dendritic spine density in mouse neurons treated with A) vehicle B)corticosterone and C) corticosterone followed by psilocybin. This demonstrates psilocybin's ability to reverse the morphological changes induced by corticosterone stress and damage, which is used here to simulate stress- and trauma-induced depression. From Zhao et al, J. Psychopharmacology, 38 (5) 2024, 489-499

2. How do we measure neuroplasticity?

Delix uses several different assays to assess neuroplasticity. One of our most useful assays has been developed with our partner Cellectricon, in which a phenotypic assay measures neurites in cultures of rat cortex cells, grown at low density and stained to highlight neurite structure. High content image analysis is then used to measure the response of neurite length and branching to treatment with test compounds as a percentage of control.

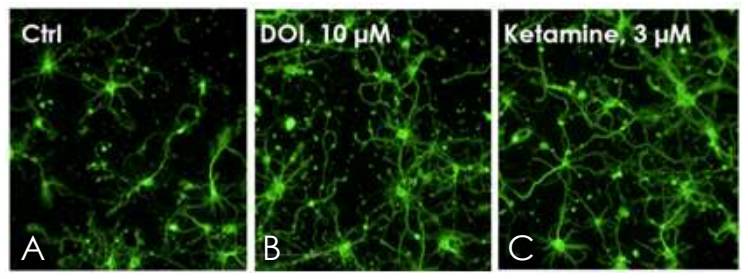
Ctrl
DOI, 10 μM
Ketamine, 3 μM
A
B
C

Representative images of rat cortex cells, stained to highlight neurite structure, after treatment with A) vehicle, B) DOI, and C) Ketamine. DOI and ketamine are hallucinogenic neuroplastogens. Courtesy Cellectricon (Mölndal, Sweden) Engel et al, Development of a high content cortical neuroplasticity assay for the assessment of structural plasticity of psychedelics. Program No 694.11 2022 Society for Neuroscience

3. How do we define actives?

The data from the phenotypic screen comes as response to treatment at a range of test compound concentrations. We postprocess the data for statistical significance and then use a variety of curve parameters to define active versus inactive compounds.

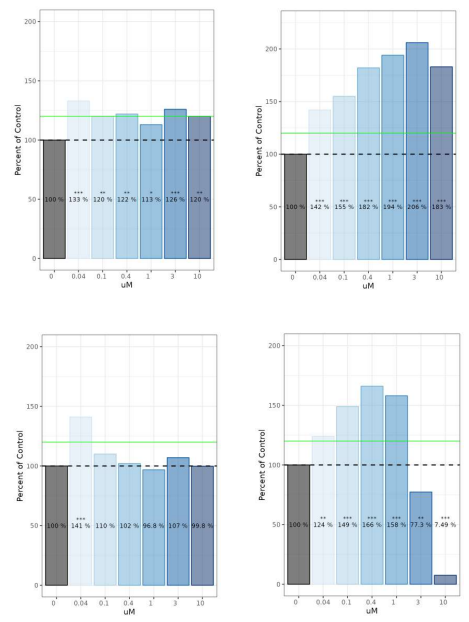

Examples of neurite response curves, showing A) Significant, flat activity across a range of concentrations B) Dose-dependent response C) A single significant response at low concentration D) an inverted U-shaped response, possibly indicating cellular toxicity or stasis at higher doses of test compound.

4. How do we build a model of neurite response?

Nearly 1,100 test compound structures and their neurite assay activities were used to create a machine learning model of neurite response, using fingerprints and molecular descriptors available within RDKit (https://www.rdkit.org) The model was created using the XGBoost method which uses gradient boosting and ensemble decision trees. The model was refined using cross validation to an internal accuracy above 0.7.

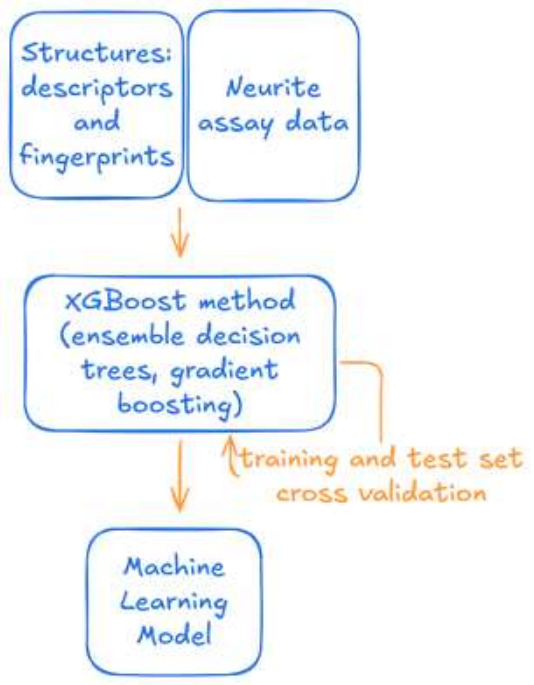
Structures: descriptors and fingerprints
Neurite assay data
XGBoost method (ensemble decision trees, gradient boosting)
training and test set cross validation
Machine Learning Model

5. How do we find novel neuroplastogens?

Structures of 752,000 commercial compounds, available for purchase as solids, were cleaned, de-salted, and filtered for reactive, undesirable, and uninteresting substructures. They were further filtered for chemical properties that were consistent with CNS active drugs. The remaining structures were then converted to molecular fingerprints and descriptors, and evaluated multiple times with the machine learning model using a range of random seeds.

Compounds that were robustly predicted to be active were then clustered. Representatives of the clusters with attractive properties like fewer chiral centers, synthetic accessibility, and CNS drug-like predicted pKa values were purchased from commercial vendors, and evaluated in the neurite assay at Cellectricon.

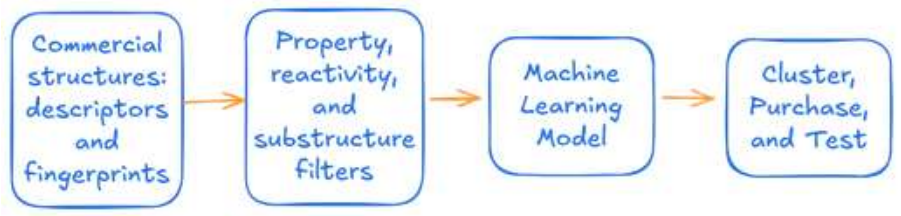
Commercial structures: descriptors and fingerprints
Property, reactivity, and substructure filters
Machine Learning Model
Cluster, Purchase, and Test

6. Did it work?

Yes, extremely well. Of the 100 compounds in this set, 65 were active in the neurite assay, compared to 44% actives in a similar exercise conducted one year earlier with fewer data. The maximum response of many of these molecules greatly exceeded the activities of hallucinogenic neuroplastogens in the same assay. These compounds have small molecular weights, logP, rotatable bonds, donors, acceptors, and polar surface areas, in the realm of known CNS-active drugs. Furthermore, they represent novel, chemically attractive structural classes distinct from the phenethylamines, tryptamines, and benzylamines of traditional hallucinogenic neuroplastogens. Several of the most active compounds have been re-synthesized and repeated their activity.

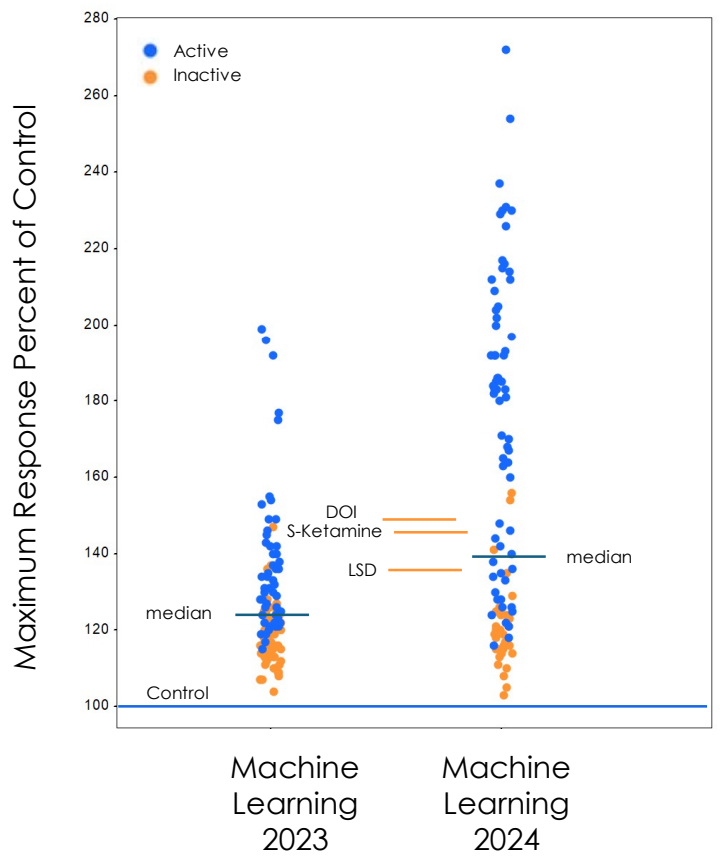
Maximum Response Percent of Control
280
260
240
220
200
180
160
140
120
100
Active
Inactive
DOI
S-Ketamine
LSD
median
median
Control
Machine Learning 2023
Machine Learning 2024